#10

Impact of HIV-1 Reverse Transcriptase E138 Mutations on Rilpivirine Drug Susceptibility

Mojgan Haddad[1], Laura A Napolitano[1], Agnes C Paquet[1], Mark C Evans[1], Christos J Petropoulos[1], Jeannette Whitcomb[1], Laurence Rimsky[2], Johan Vingerhoets[2], Gaston Picchio[3], Eoin Coakley[1]

[1]Monogram Biosciences, South San Francisco, CA, USA; [2]Tibotec, Beerse, Belgium, [3]Tibotec, Titusville, NJ

Monogram Biosciences, Inc.
345 Oyster Point Blvd.
South San Francisco, CA 94080
Lnapolitano@monogrambio.com

International HIV&HEPATITIS VIRUS Drug Resistance Workshop & Curative Strategies

Los Cabos, Mexico. June 8-11, 2011

BACKGROUND

Rilpivirine (RPV) is a recently approved non-nucleoside reverse transcriptase inhibitor (NNRTI) that has shown effective virologic response in two Phase III studies (ECHO and THRIVE) of treatment naïve individuals. Analysis of RPV virologic failure specimens has identified emergence of NNRTI resistance-associated mutations, particularly at the reverse transcriptase E138 codon. In this study we implemented stepwise approach to:

1) Determine the upper confidence limit of the distribution of phenotypic RPV susceptibility, i.e. the RPV biological cutoff (BCO) in a phenotypic assay

2) Define the levels of RPV susceptibility associated with differing substitutions at RT codon 138.

METHODS

A multi-step approach was taken:

- The BCO was defined as the RPV fold change (FC) in IC50 below which reside 99% of the samples in this distribution

- Initial BCO was determined in a population of clinical samples submitted for combination phenotypic and genotypic testing that had no typical NNRTI, NRTI or PI RAMS (the reference wild-type population).
 - NRTI RAMs controlled for (excluded): M41L, K65R, D67N, T69, K70R, K70E, L74, V75A/M/S/T, Y115F, Q151M, M184, L210W, T215F/Y, K219
 - NNRTI RAMs controlled for (excluded): A98G, L100I, **K101E**, **K101P**, K103N, K103S, V106A, V106M, **Y181**, Y188, G190, P225, F227, M230L, P236L
 - PI RAMs controlled for (excluded): L24, D30, V32, M46, I47, G48, I50, I54, V82A, V82, I84, N88, L90

- Next, established RPV mutations were excluded from the wildtype reference population
 - Phenotypic distributions of viruses with E138A, G, K, or Q mutations were each examined in isolation. There were insufficient samples with E138R for this analysis.

- Mann-Whitney test was performed to evaluate the individual impact of each of those mutations on the FC distribution.

RESULTS

- 7,257 wildtype subtype B specimens were studied. All specimens had matched genotype and phenotype with RPV FC. In this full data set the BCO was 2.5. **(Figure 1)**

- Exclusion of samples with RPV RAMs E138G,K,Q,R and H221Y lowered the BCO to 2.4 **(Figure 2)**

- Exclusion of samples with E138A further reduced the BCO to 2.0. **(Figure 3)**

- Median FC of samples (and % of wildtype reference population) with E138A, G, K, and Q were 1.9 (2.7%), 2.7 (0.055%), 1.5 (0.12%) and 2.6 (0.055%), respectively. **(Figures 4 and 5)**

- The FC distributions of these samples were statistically significantly higher than samples without these mutations (p-value < 0.05). **(Figure 4)**

Figure 1: Biological Cutoff: Wildtype Dataset
Excluded: traditional PI, NRTI, NNRTI RAMs

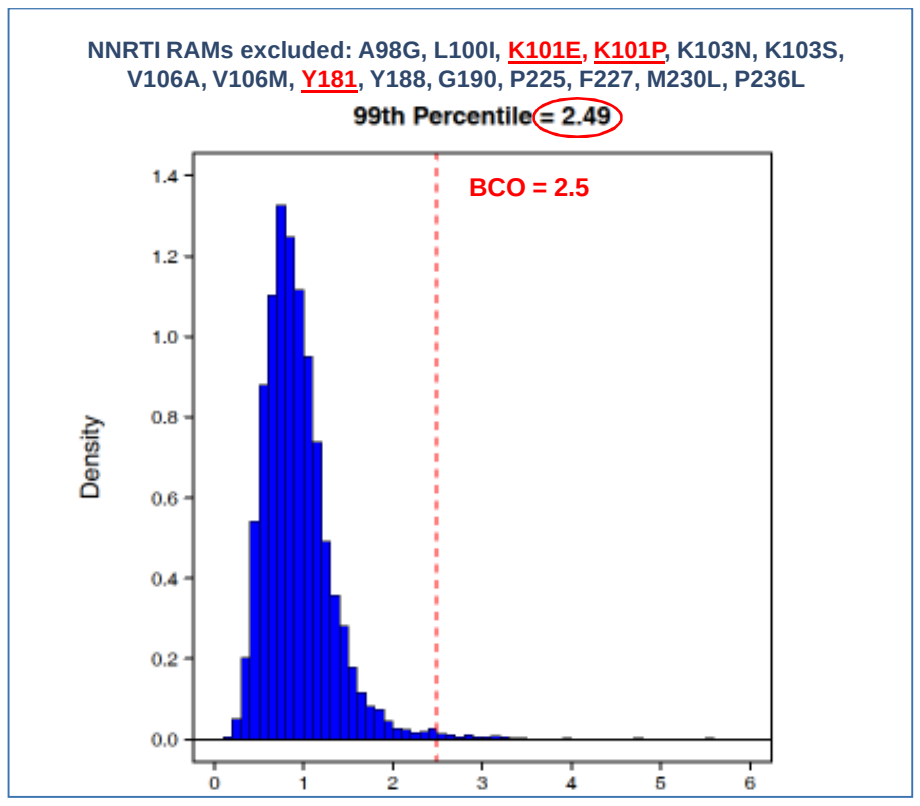
NNRTI RAMs excluded: A98G, L100I, K101E, K101P, K103N, K103S, V106A, V106M, Y181, Y188, G190, P225, F227, M230L, P236L
99th Percentile = 2.49
BCO = 2.5
Density

Figure 2: Biological Cutoff: Wildtype Dataset
Exclude Established RPV RAMs

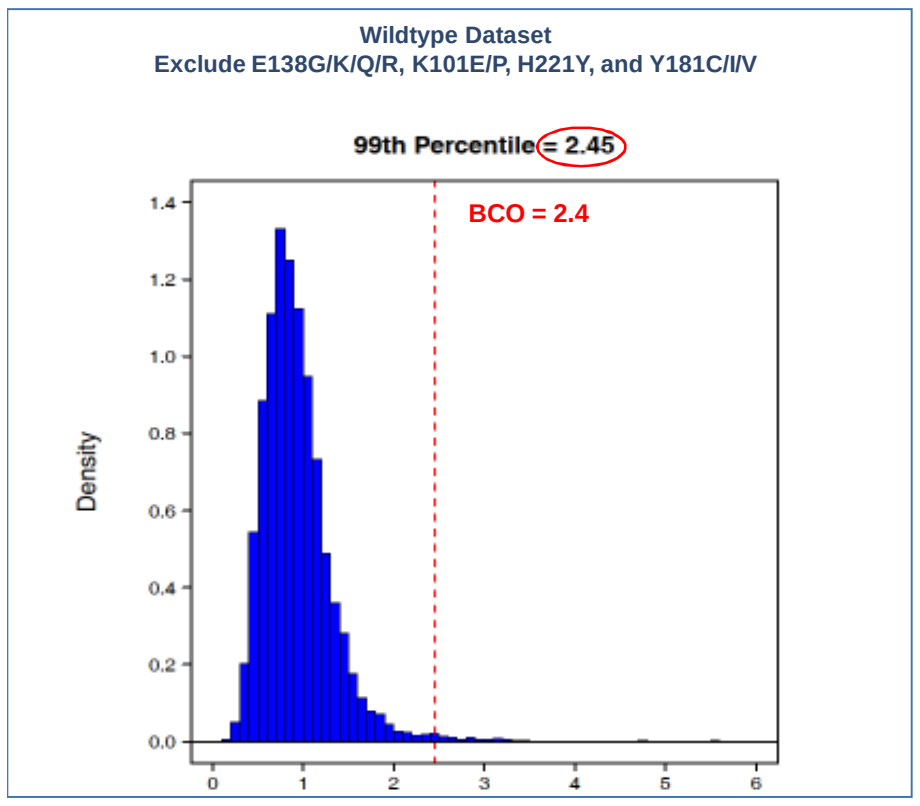
Wildtype Dataset
Exclude E138G/K/Q/R, K101E/P, H221Y, and Y181C/I/V
99th Percentile = 2.45
BCO = 2.4
Density

Figure 3 Biological Cutoff: Wildtype Dataset
Exclude Established RPV RAMs and E138A

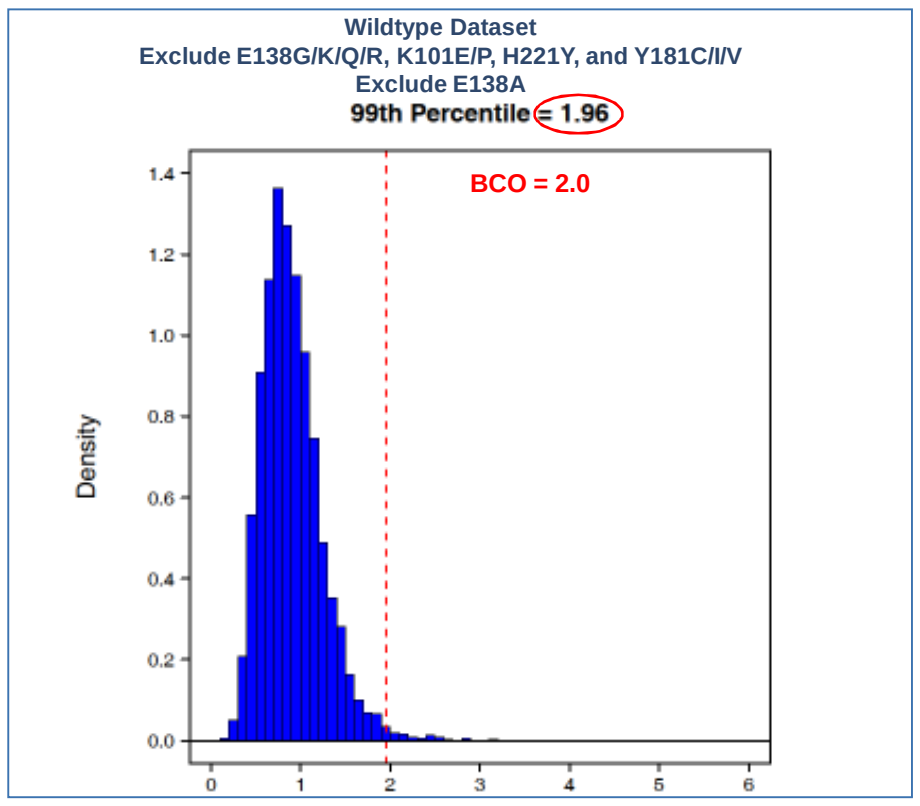
Wildtype Dataset
Exclude E138G/K/Q/R, K101E/P, H221Y, and Y181C/I/V
Exclude E138A
99th Percentile = 1.96
BCO = 2.0
Density

Figure 4: Impact of E138 Mutations on RPV Fold-Change
Comparison of RPV FC in Wildtype With and Without E138 Mutations

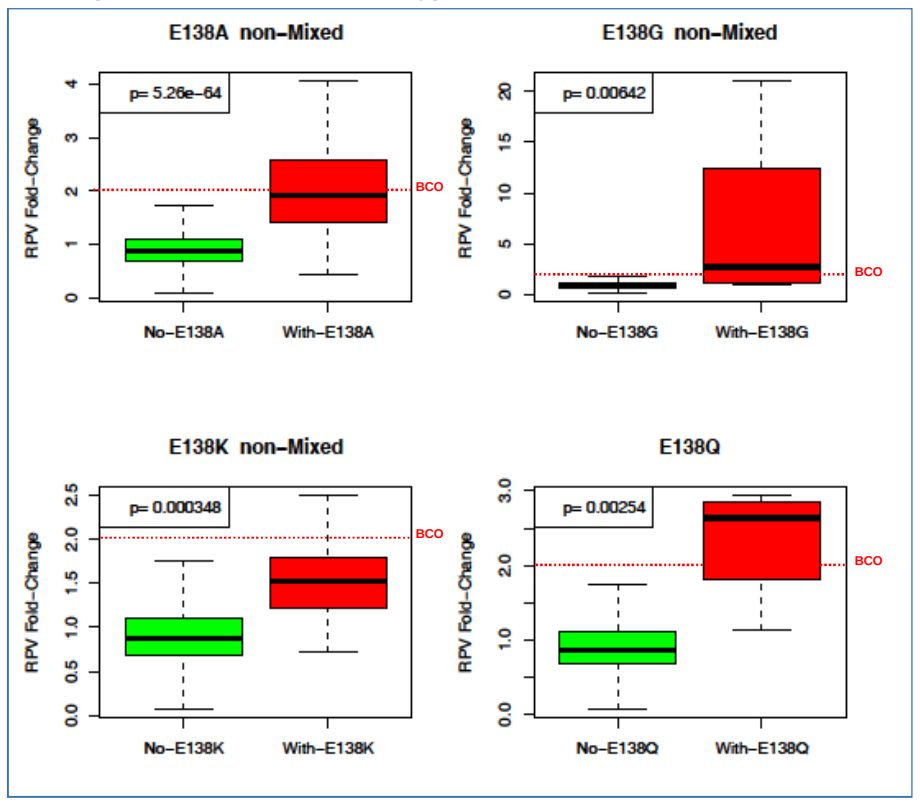
E138A non–Mixed
p= 5.26e–64
BCO
No–E138A
With–E138A
E138G non–Mixed
p= 0.00642
No–E138G
With–E138G
E138K non–Mixed
p= 0.000348
No–E138K
With–E138K
E138Q
p= 0.00254
No–E138Q
With–E138Q
RPV Fold–Change

Figure 5: Impact of E138 Mutations on RPV Fold-Change

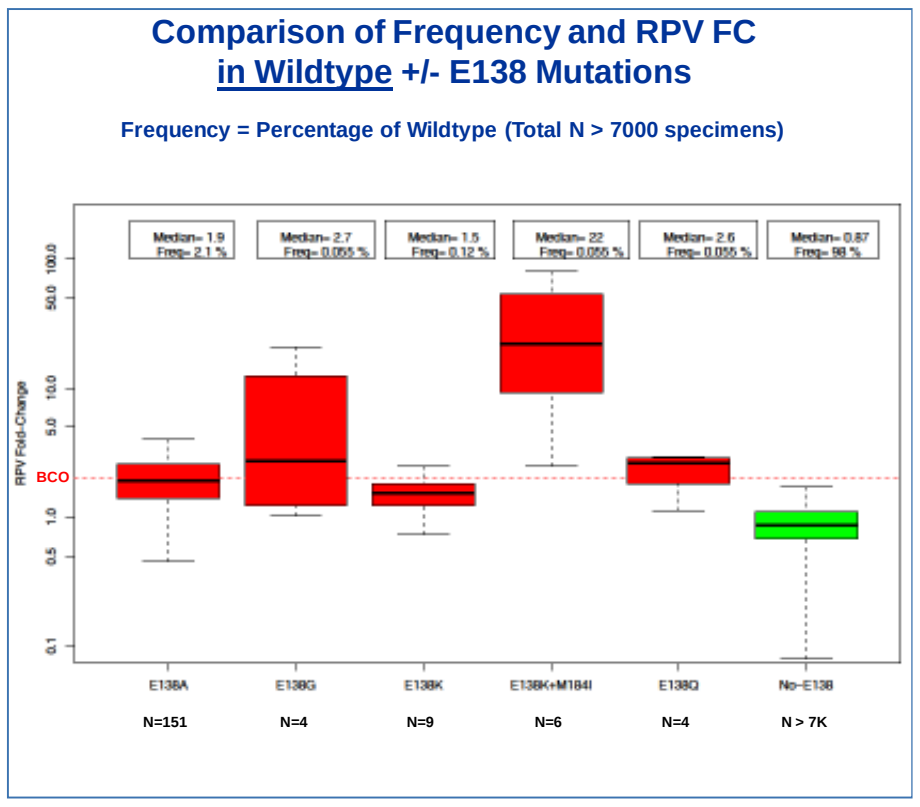
Comparison of Frequency and RPV FC
in Wildtype +/- E138 Mutations
Frequency = Percentage of Wildtype (Total N > 7000 specimens)
RPV Fold-Change
BCO
E138A
E138G
E138K
E138K+M184I
E138Q
No-E138
N=151
N=4
N=9
N=6
N=4
N > 7K

CONCLUSIONS

- Excluding E138A from the wildtype reference population establishes the BCO at 2.0 in a phenotypic assay.

- E138A confers resistance similar to E138G/K/Q, though with lower range in FC relative to G & Q .

- Recent FDA prescribing information identifies E138A as an RPV RAM (with V179L, F227C, M230I/L).

- Continued correlative analyses of RPV genotypic and phenotypic resistance data is vital to the development and accuracy of interpretative algorithms

ACKNOWLEDGEMENTS: The authors thank the Monogram Biosciences Clinical Reference Laboratory for performance of all assays.